UNIVERSITÀ DEGLI STUDI DI MILANO
DIPARTIMENTO DI SCIENZE FARMACOLOGICHE
E BIOMOLECOLARI - DiSFeB

65th Annual Meeting of the
Society for Medicinal Plant and Natural Product Research
Basel, Switzerland,
3rd - 7th September 2017

Anti-inflammatory and antioxidant activities of two different **Ginkgo biloba L.** extracts in human endothelial cells: a comparative study

Stefano Piazza[1], Barbara Pacchetti[2], Marco Fumagalli[1], Enrico Sangiovanni[1], Mario Dell'Agli[1]

[1] Department of Pharmacological and Biomolecular Sciences, Università degli Studi di Milano, Milano, Italy
[2] Linnea SA, Riazzino, Switzerland

Cardiovascular and cerebrovascular dysfunctions are widespread in developed country as a consequence of ageing and lifestyle. Extracts from leaves of **Ginkgo biloba L.** are widely used for their beneficial effects in cardiovascular diseases prevention and therapy or to improve mild cognitive impairment and cerebrovascular insufficiency. Studies in the literature clearly demonstrate that an adequate standardization of the extracts, that should contain flavone glycosides (22.0-27.0%) and terpene lactones (5.0-7.0%) including ginkgolides A, B and C, and bilobalide (ESCOP, 2009), is mandatory for efficacy. Standardized extracts are prepared by solvents extraction. Despite extraction with acetone is reported by European Pharmacopoeia and acetone extract is the most clinically investigated (EGb761®), in some countries the amount of solvent allowed in food supplements is limited to ethanol, thus implying that the usage of a standardized Ginkgo b.ethanol extract is preferred.

Aim of the work

The aim of the present study was to 1) chemically profile acetone (G24) vs. ethanol (G4E) extracts from Ginkgo biloba leaves similarly standardized to identify differences among components; 2) to investigate if the extracts may be considered comparable as anti-inflammatory and antioxidant agents.

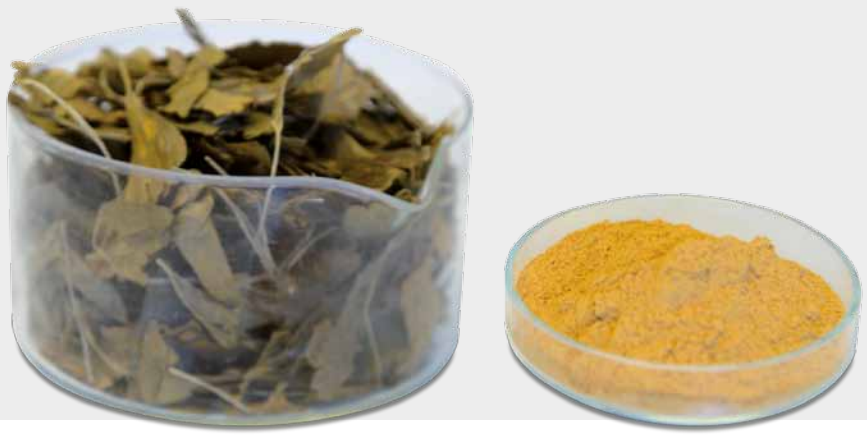

Methods

CELL CULTURE
Human endothelial cell (HUVEC, CRL-1730, ATCC) were grown in Ham F12K medium and treated at passage number 2-30 with the indicated concentration of *Ginkgo biloba L.* extracts or vehicle alone (<0.2% DMSO). Curcumin at 10 μM was used as reference compound.

PLANT MATERIAL
Ginkgo leaves were collected by plantations cultivated in Europe and North America under controlled conditions. **G4E and G24 powdered extracts were provided by Linnea SA** (Riazzino, Locarno, CH). Patented standardized extraction and purification processes were applied to obtained extracts with consistent composition in pharmacologically active compounds.

CYTOTOXICITY ASSAY
The cytotoxicity of the extracts was evaluated by the 3,4,5-dimethythiazol-2-yl-5-diphenyltetrazolium bromide (MTT) assay. No cytotoxicity at concentrations used in the present study was observed.

MEASUREMENT OF SOLUBLE ADHESION MOLECULES
For the measurement of soluble ICAM-1, VCAM-1 and E-selectin adhesion molecules, HUVEC cells were grown in 24-wells plates (20.000 cells/well) for 72 h, then treated with the pro-inflammatory stimulus (TNFα at 20 ng/mL) and the extracts under study for 6 h. Soluble adhesion molecules were quantified in the cells media by an enzyme-linked immunosorbent assay (ELISA).

MEASUREMENT OF ROS PRODUCTION
To evaluate the effect of the extracts on ROS formation, HUVEC were grown in black 96-wells plate (10.000 cells/well) for 72 h, and then treated with the extracts for 24 h. After the treatment, cells were incubated for 1 h with a fluorescent probe (CM-H2DCFDA) and then washed with PBS 1X before the pro-oxidant stimulation with H_2O_2 (1 mM for 1 h).
The fluorescence of the internalized probe was read, after a final wash with PBS, by microplate reader at 535 nm. Trolox (500 μM) was used as reference compound.

NF-KB NUCLEAR TRANSLOCATION
To assess the effect of the extracts on the NF-κB (p65) nuclear translocation, HUVEC were plated in 100 mm plates (5 x 10^5 cells) for 72 h, cells were treated for 1 h with the pro-inflammatory mediator and the extracts under study. After quantification by the Bradford method (Bio-Rad), 10 μg/well of nuclear proteins (Nuclear Extraction Kit from Cayman Chemical company) were assayed by ELISA. NF-κB translocation was measured by spectroscopy at 450 nm.

Linnea
Natural Pharma Solutions

Results and discussion

CHARACTERIZATION OF GINKGO BILOBA EXTRACTS

The extracts (20 batches each) G4E and G24 (*table1*) showed the same profile in terms of proanthocyanidins, bilobalide, and flavone glycosides content. According to these results, the major classes of constituents occurring in *Ginkgo biloba* acetone extract, which is considered the best extract to provide biological activity, are present in the ethanol G4E extract at comparable concentrations. Standardization was applied as follows: 24% flavone glycosides and 6.0% terpene lactones for both G24 and G4E.

TABLE 1.
CHARACTERIZATION OF G24 AND G4E EXTRACTS

	G24 EXTRACT % (mean ± s.d.)	G4E EXTRACT
BILOBALIDE	2.94 ± 0.13	2.87 ± 0.13
PROANTHOCYANIDINS	3.62 ± 2.65	4.81 ± 2.42
FLAVONE GLYCOSIDES	25.31 ± 1.04	25.03 ± 0.94

G4E AND G24 EXTRACTS INHIBIT ADHESION MOLECULES RELEASE

Either of G4E and G24 the extracts inhibited in a concentration dependent way soluble adhesion molecules VCAM-1, ICAM-1 and E-selectin release (*figure 1*) induced by TNFα, with similar IC_{50} (*table 2*). The effect was statistically significant at 50 μg/ml already. Both the extracts showed higher activity in inhibiting VCAM-1 release, while the inhibitory effect on ICAM-1 and E-selectin was less pronounced (*table 2*). Chen et al. demonstrated inhibition of adhesion molecules by a *Ginkgo biloba* standardized extract provided by Dr. Schwabe (Chen et al., 2003), which is considered comparable to our acetone extract.
Although studies report that variability may occur within different formulation, ginkgolides and flavonols could reach significant systemic concentrations, suggesting VCAM-1 release could be inhibited in vivo after consumption of standardized extracts.

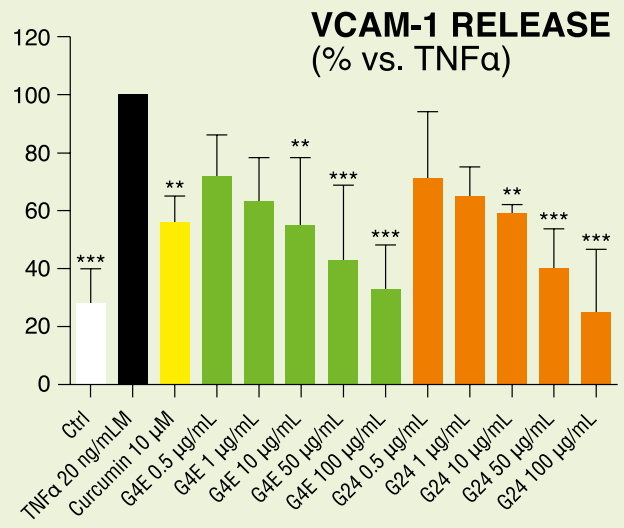

VCAM-1 RELEASE
(% vs. TNFα)
0
20
40
60
80
100
120
Ctrl
TNFα 20 ng/mLM
Curcumin 10 μM
G4E 0.5 μg/mL
G4E 1 μg/mL
G4E 10 μg/mL
G4E 50 μg/mL
G4E 100 μg/mL
G24 0.5 μg/mL
G24 1 μg/mL
G24 10 μg/mL
G24 50 μg/mL
G24 100 μg/mL

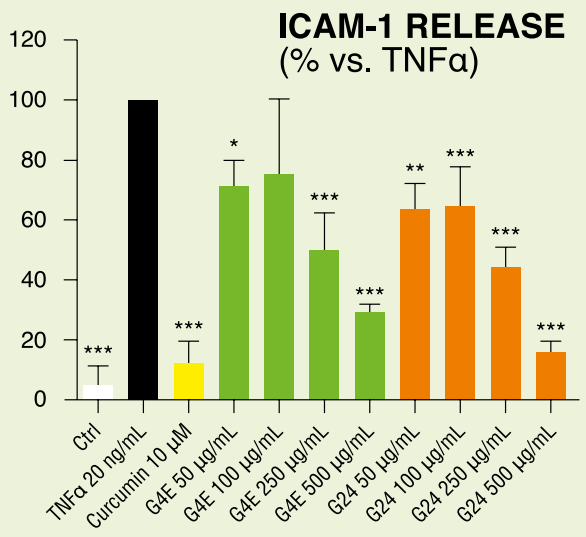

ICAM-1 RELEASE
(% vs. TNFα)
0
20
40
60
80
100
120
Ctrl
TNFα 20 ng/mL
Curcumin 10 μM
G4E 50 μg/mL
G4E 100 μg/mL
G4E 250 μg/mL
G4E 500 μg/mL
G24 50 μg/mL
G24 100 μg/mL
G24 250 μg/mL
G24 500 μg/mL

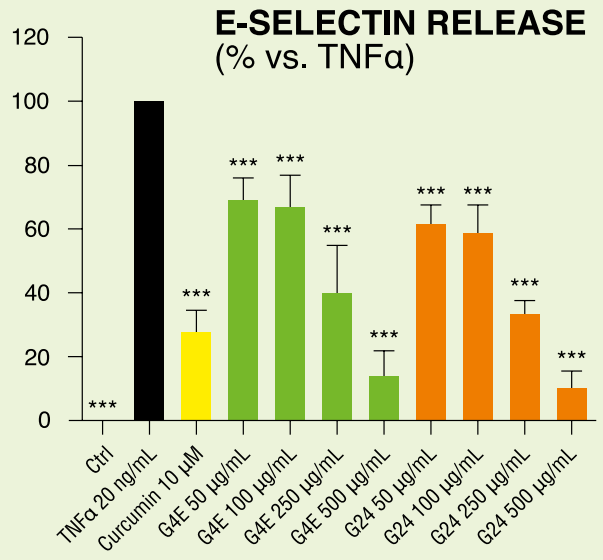

E-SELECTIN RELEASE
(% vs. TNFα)
0
20
40
60
80
100
120
Ctrl
TNFα 20 ng/mL
Curcumin 10 μM
G4E 50 μg/mL
G4E 100 μg/mL
G4E 250 μg/mL
G4E 500 μg/mL
G24 50 μg/mL
G24 100 μg/mL
G24 250 μg/mL
G24 500 μg/mL

G4E AND G24 EXTRACTS INHIBIT ROS PRODUCTION IN HUMAN ENDOTHELIAL CELLS

ROS formation was inhibited by G4E and G24 in a concentration dependent fashion *(figure 2*), with similar IC50 (*table 2*). Our data agree with those previously published reporting reduction of ROS production and oxidative stress although in different *Ginkgo biloba* extracts and cell models. However, this is the first report demonstrating that the inhibitory effect on ROS formation in human endothelial cells by G4E and G24 is comparable.

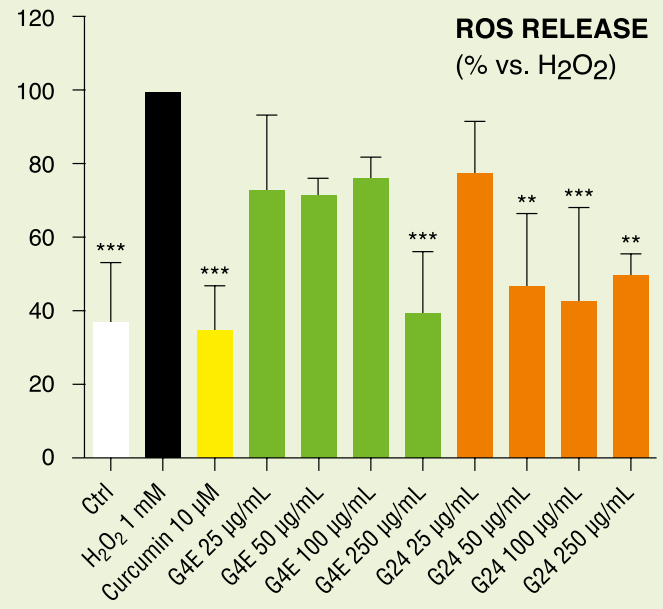

ROS RELEASE
(% vs. H_2O_2)
0
20
40
60
80
100
120
Ctrl
H_2O_2 1 mM
Curcumin 10 μM
G4E 25 μg/mL
G4E 50 μg/mL
G4E 100 μg/mL
G4E 250 μg/mL
G24 25 μg/mL
G24 50 μg/mL
G24 100 μg/mL
G24 250 μg/mL

TABLE 2.
IC50 (μg/ml) OF GINKGO EXTRACTS FOR THE TESTED BIOLOGICAL ACTIVITIES

	G24	G4E
VCAM-1	1.52	1.50
ICAM-1	129	209
E-SELECTIN	137	170
ROS FORMATION	38.8	57.6
NF-KB TRANSLOCATION	77.5	70.4

G4E AND G24 INHIBIT NF-κB NUCLEAR TRANSLOCATION IN HUMAN ENDOTHELIAL CELLS

NF-κB is known as inflammatory and redox sensitive transcription factor activated by ROS and cytokines such as TNFα, which has a key role in the expression of adhesion molecules by human endothelial cells. To evaluate if the extracts could impair NF-κB activation, HUVEC were treated with G4E and G24 (20-250 μg/ml) for 1 hour. Both the extracts inhibited NF-κB translocation in a concentration dependent fashion (*figure 3*), with comparable IC_{50} (*table 2*).

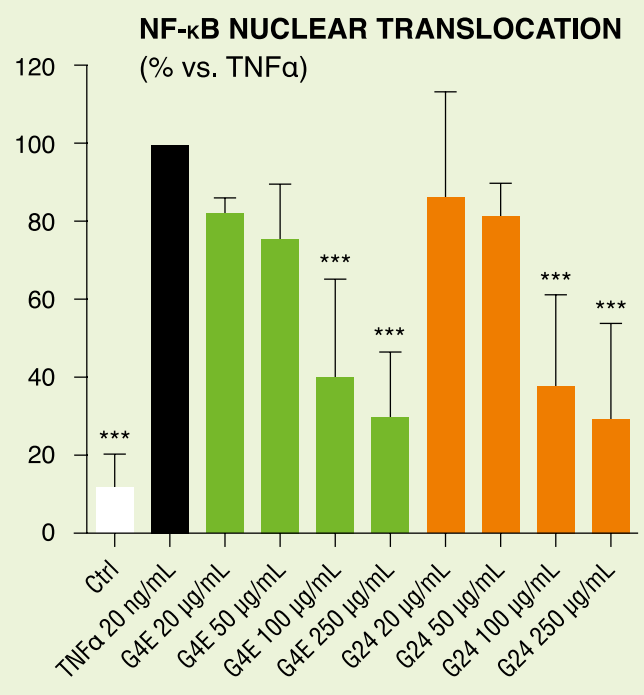

NF-κB NUCLEAR TRANSLOCATION
(% vs. TNFα)
0
20
40
60
80
100
120
Ctrl
TNFα 20 ng/mL
G4E 20 μg/mL
G4E 50 μg/mL
G4E 100 μg/mL
G4E 250 μg/mL
G24 20 μg/mL
G24 50 μg/mL
G24 100 μg/mL
G24 250 μg/mL

Conclusions

Although European Pharmacopoeia recommends extraction of *Ginkgo* powder with acetone, the present paper demonstrates for the first time that ethanol (G4E) and acetone (G24) extracts show comparable anti-inflammatory and antioxidant activity in human endothelial cells. These results provide new insights on the usage of ethanol extracts in those countries where restrictions in amount of acetone in food supplements are present.

Grant
The research was supported with contribution of Linnea SA, Switzerland

LINNEA SA
Via Cantonale, CH-6595 Riazzino (TI), Switzerland
Tel. +41 (0)91 850 5050 - Email:info@linnea.ch

Natural Pharma Solutions
www.linnea.ch